Development of ^{212}Pb-based Radio-DARPin therapy (RDT) for the treatment of mesothelin (MSLN)-positive solid tumors

S Wullschleger[1], A Saidi[2], D Buffet[1], T Stallons[2], S Riesenberg[1], A Wong[2], M Paladino[1], M Müller[1], C Herzog[1], J Walter[1], D Villemagne[1], Y Kaufmann[1], N Pina[1], H Fernandez[1], T Lekishvili[1], E Tselempi[1], J Blunschi[1], L Abduli[1], C Iss[1], A Croset[1], C Domke[1], A Schatzmann[2], A Goubier[1], J Torgue[2], D Steiner[1]

1. Molecular Partners AG, Zurich-Schlieren, Switzerland, 2. Orano Med LLC, Plano, TX, USA

Poster/abstract #339

2025 AACR Annual Meeting

Introduction

MSLN is an attractive target for ovarian cancer

- MSLN is expressed in ovarian cancer with high prevalence (> 80%) and high level of intra-tumoral homogeneity[1]
- Expression is maintained in lymph node and peritoneal metastases[1]
- MSLN expression in healthy tissue is primarily restricted to mesothelial cells

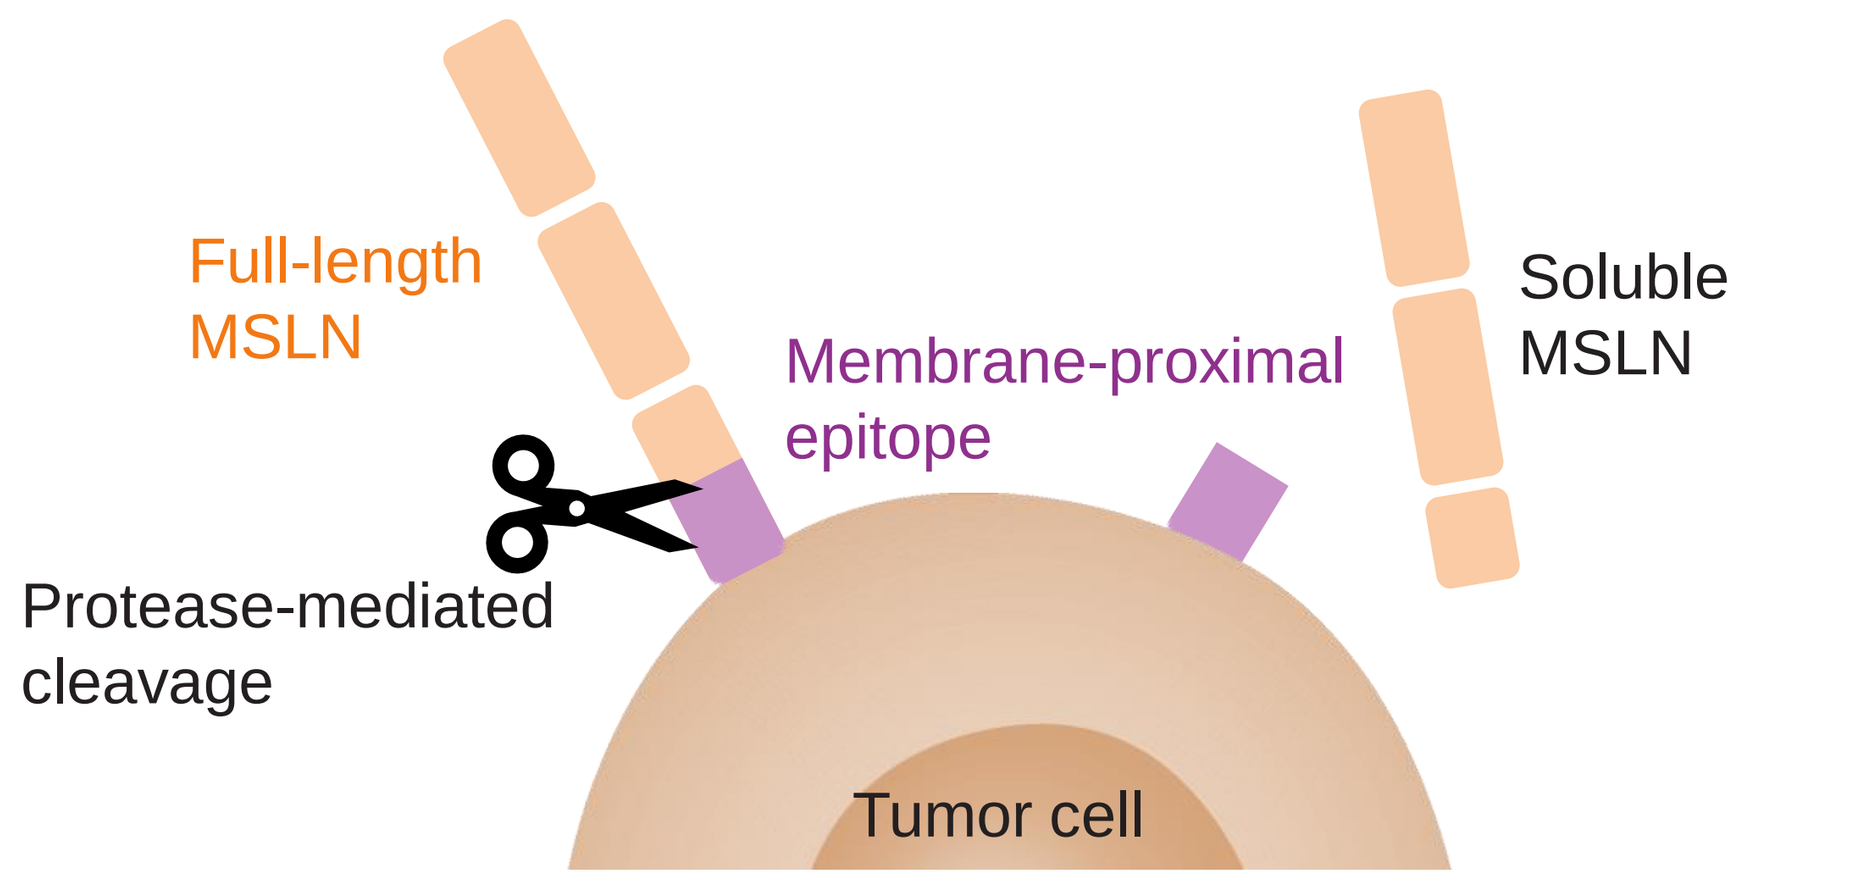
Full-length MSLN
Membrane-proximal epitope
Soluble MSLN
Protease-mediated cleavage
Tumor cell

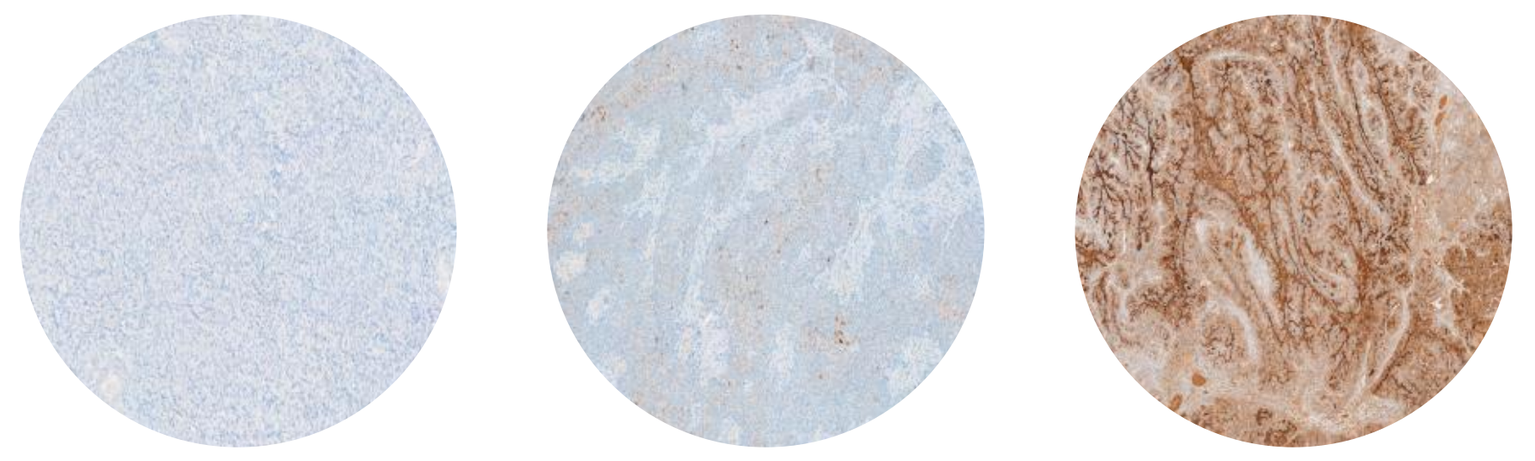

MSLN IHC staining pattern on normal ovarian tissue (left) and ovarian tumors: moderate (middle) and strong (right) expression.

- MSLN is a GPI-anchored surface protein
- MSLN is cleaved by multiple proteases in its membrane-proximal region
- Systemic soluble form (sMSLN) up to 160 ng/ml measured in ovarian cancer patients[2, 3]
- sMSLN can sequester anti-MSLN therapeutics and potentially diminish their efficacy

^{212}Pb x MSLN RDT

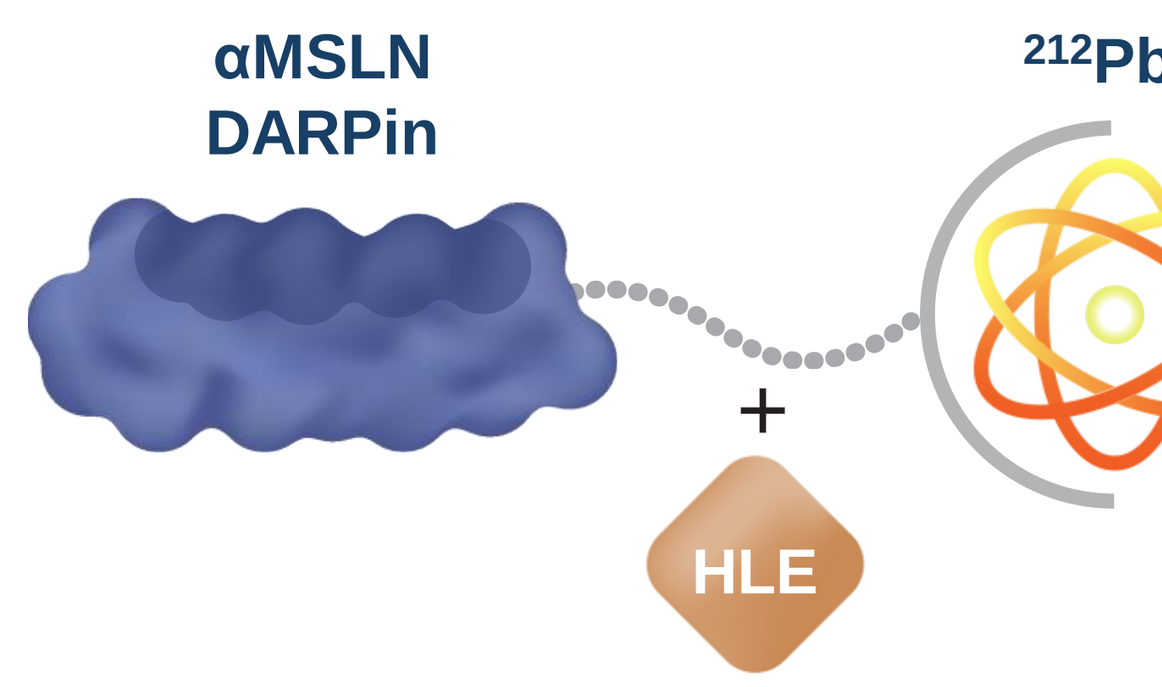
αMSLN DARPin
212Pb
+
HLE

- Designed Ankyrin Repeat Protein (DARPin) that targets a membrane-proximal epitope of MSLN was selected to prevent interaction with sMSLN (HLE: half-life extending moiety)
- The radioactive payload is ^{212}Pb, an alpha particle-emitting radionuclide with a short decay half-life which allows high energy deposition on tumor in a short time frame

DARPins bind to MSLN with high affinity

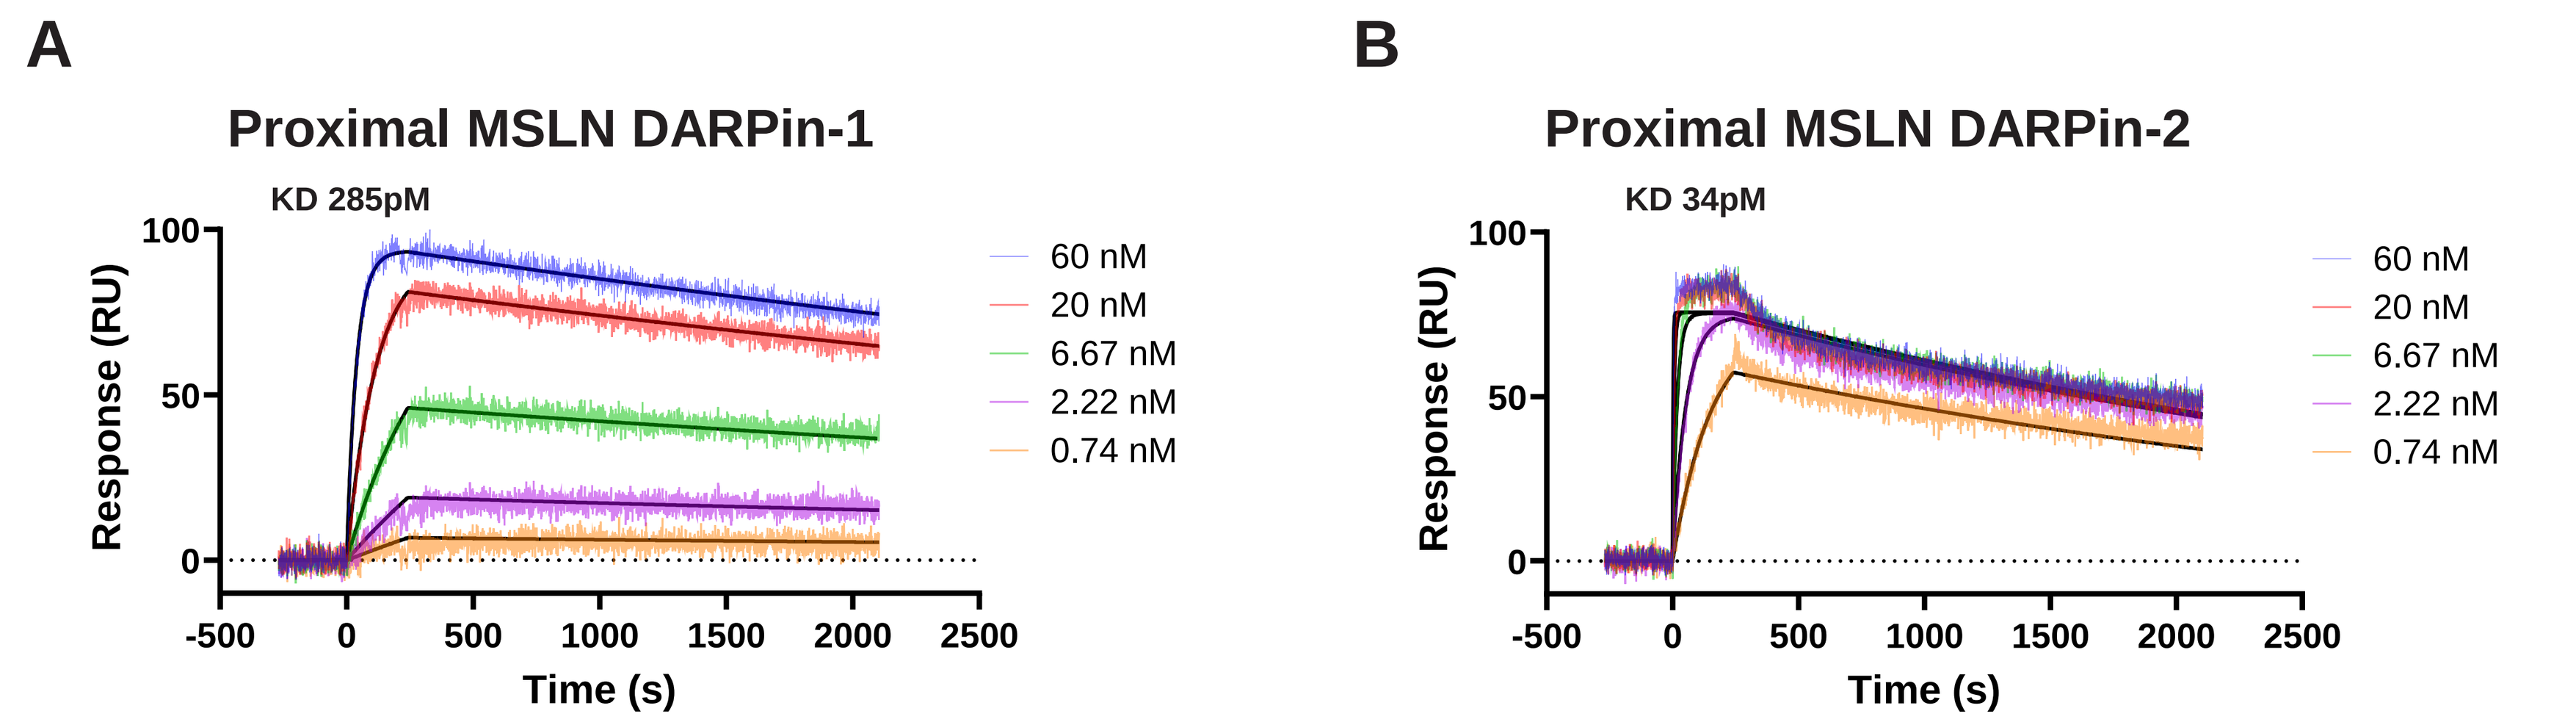
A
Proximal MSLN DARPin-1
KD 285pM
Response (RU)
Time (s)
60 nM
20 nM
6.67 nM
2.22 nM
0.74 nM
B
Proximal MSLN DARPin-2
KD 34pM
Response (RU)
Time (s)
60 nM
20 nM
6.67 nM
2.22 nM
0.74 nM

Figure 1: DARPin binding by Surface Plasmon Resonance (SPR)
DARPin binding to MSLN was assessed by SPR. A) SPR sensorgram of Proximal MSLN DARPin-1 x HLE (KD= 285 pM). B) SPR sensorgram of Proximal MSLN DARPin-2 x HLE (KD= 34 pM).

Proximal MSLN DARPins bind to membrane proximal domain of MSLN

DARPin epitope mapping by ELISA

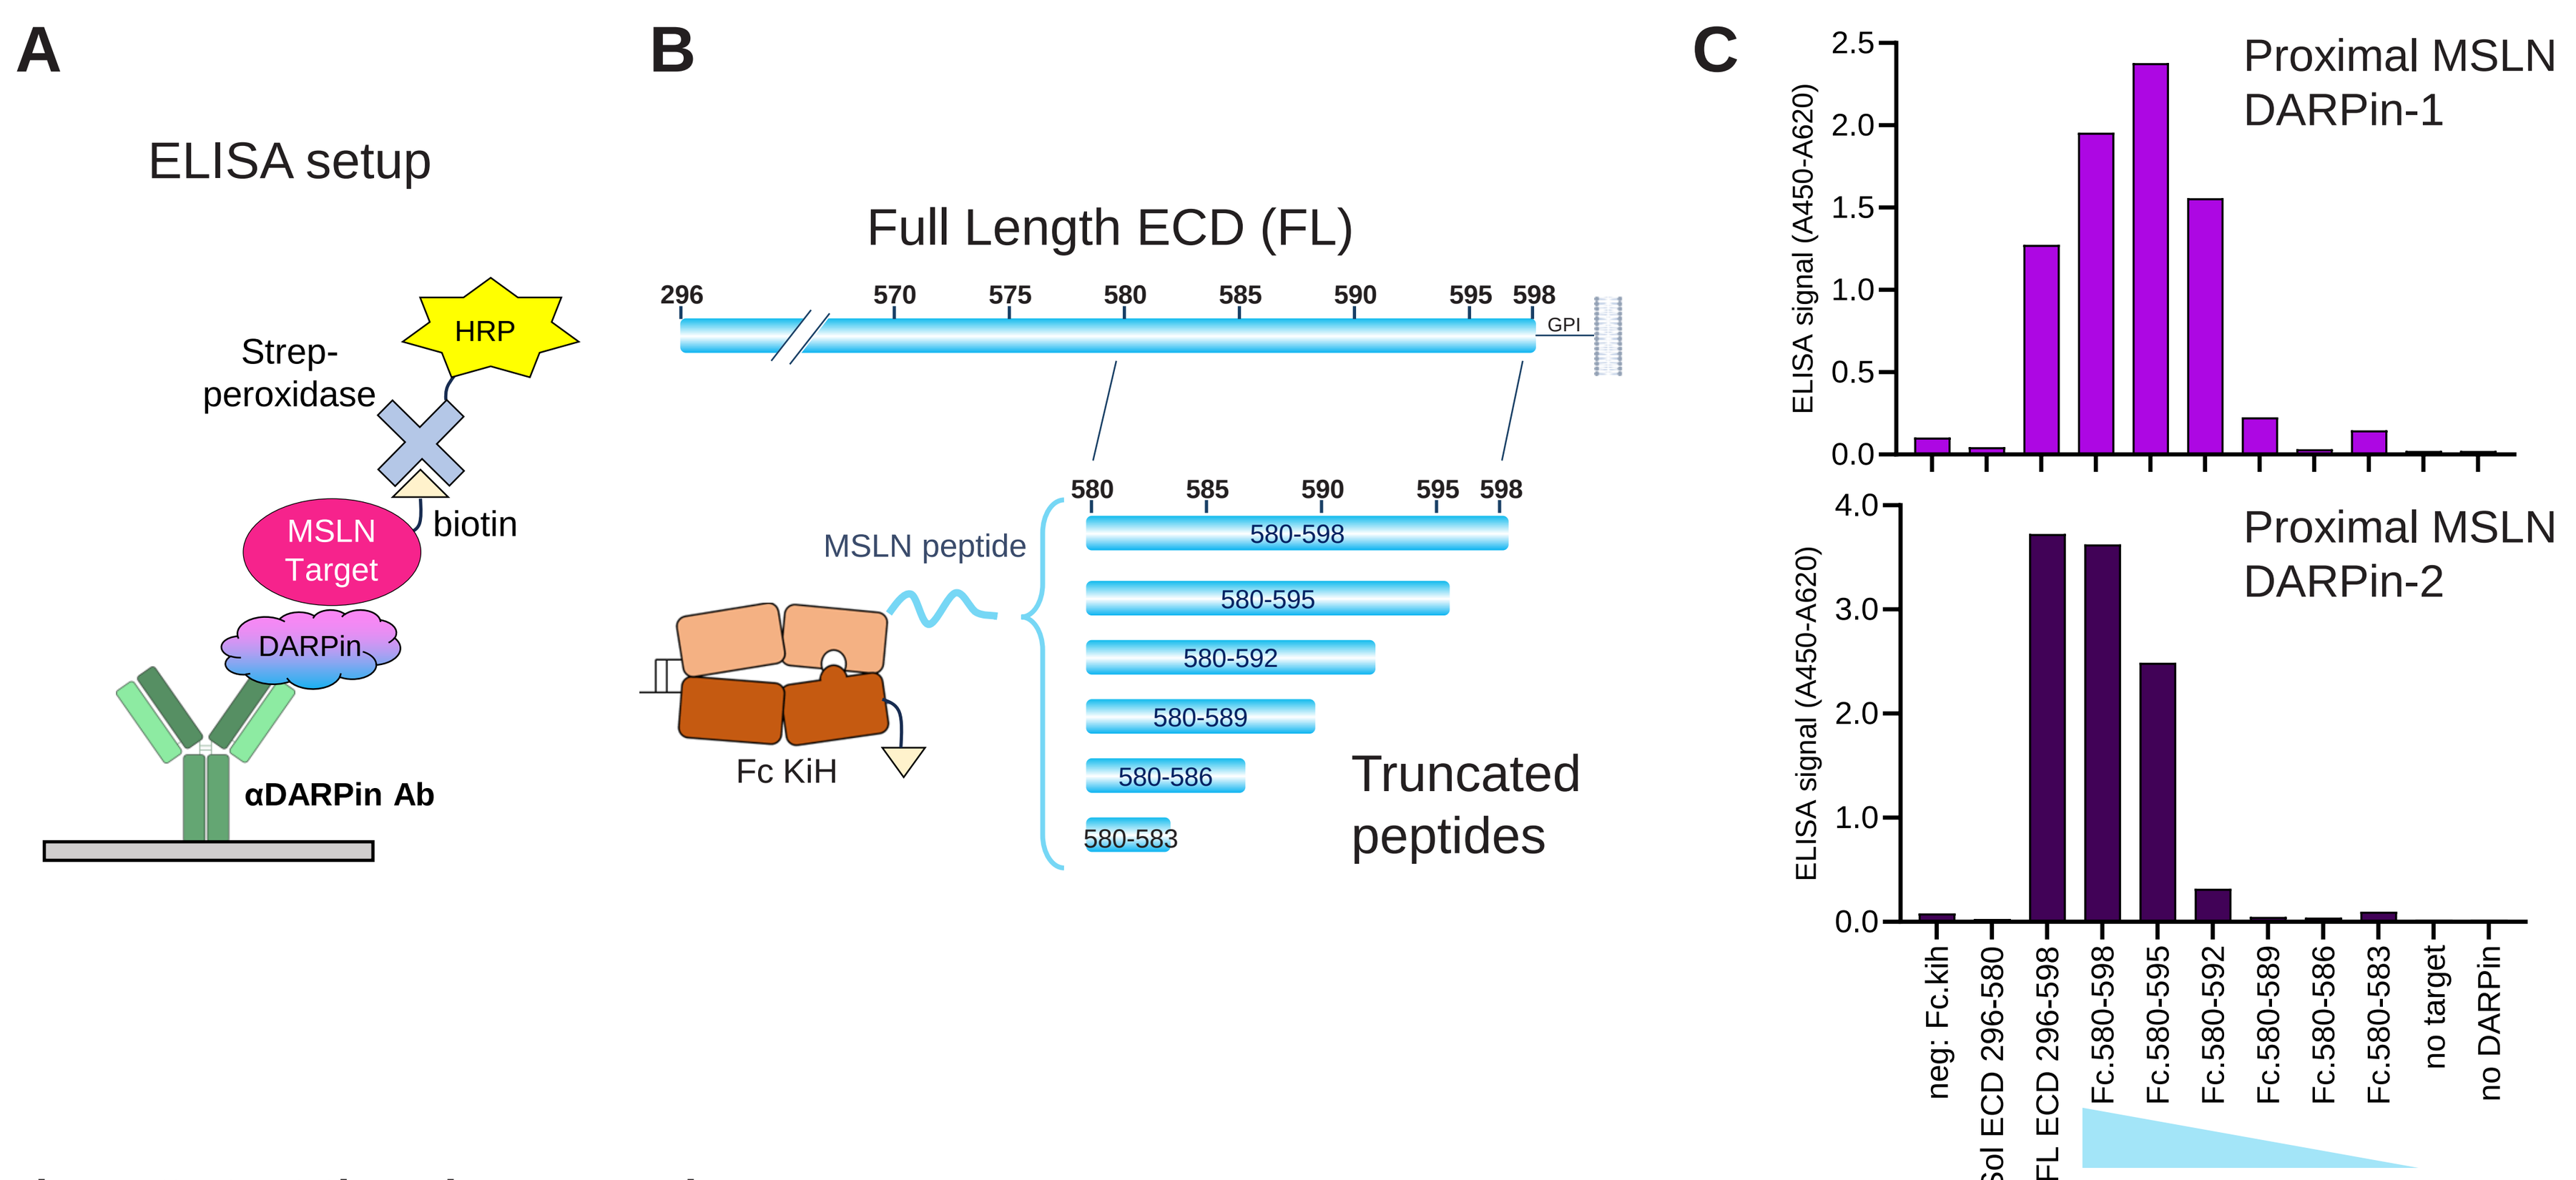
A
ELISA setup
Strep-peroxidase
HRP
biotin
MSLN Target
DARPin
αDARPin Ab
B
Full Length ECD (FL)
296
570
575
580
585
590
595
598
GPI
MSLN peptide
Fc KiH
580-598
580-595
580-592
580-589
580-586
580-583
Truncated peptides
C
Proximal MSLN DARPin-1
Proximal MSLN DARPin-2
ELISA signal (A450-A620)
neg: Fc.kih
Sol ECD 296-580
FL ECD 296-598
Fc.580-598
Fc.580-595
Fc.580-592
Fc.580-589
Fc.580-586
Fc.580-583
no target
no DARPin

Figure 2: DARPin epitope mapping
A) Experimental set up of the ELISA. DARPins are captured with an anti-DARPin antibody and the binding to MSLN target material is tested. B) Schematic representation of the MSLN target material, consisting of the full length ectodomain (ECD) or Fc construct containing truncated MSLN peptides. C) MSLN epitope mapping data for two MSLN DARPins. Fc construct only, soluble ECD, no target and no DARPin were used as controls.

Cell binding of proximal MSLN DARPins is not affected by sMSLN

Cell binding of DARPins in presence of sMSLN

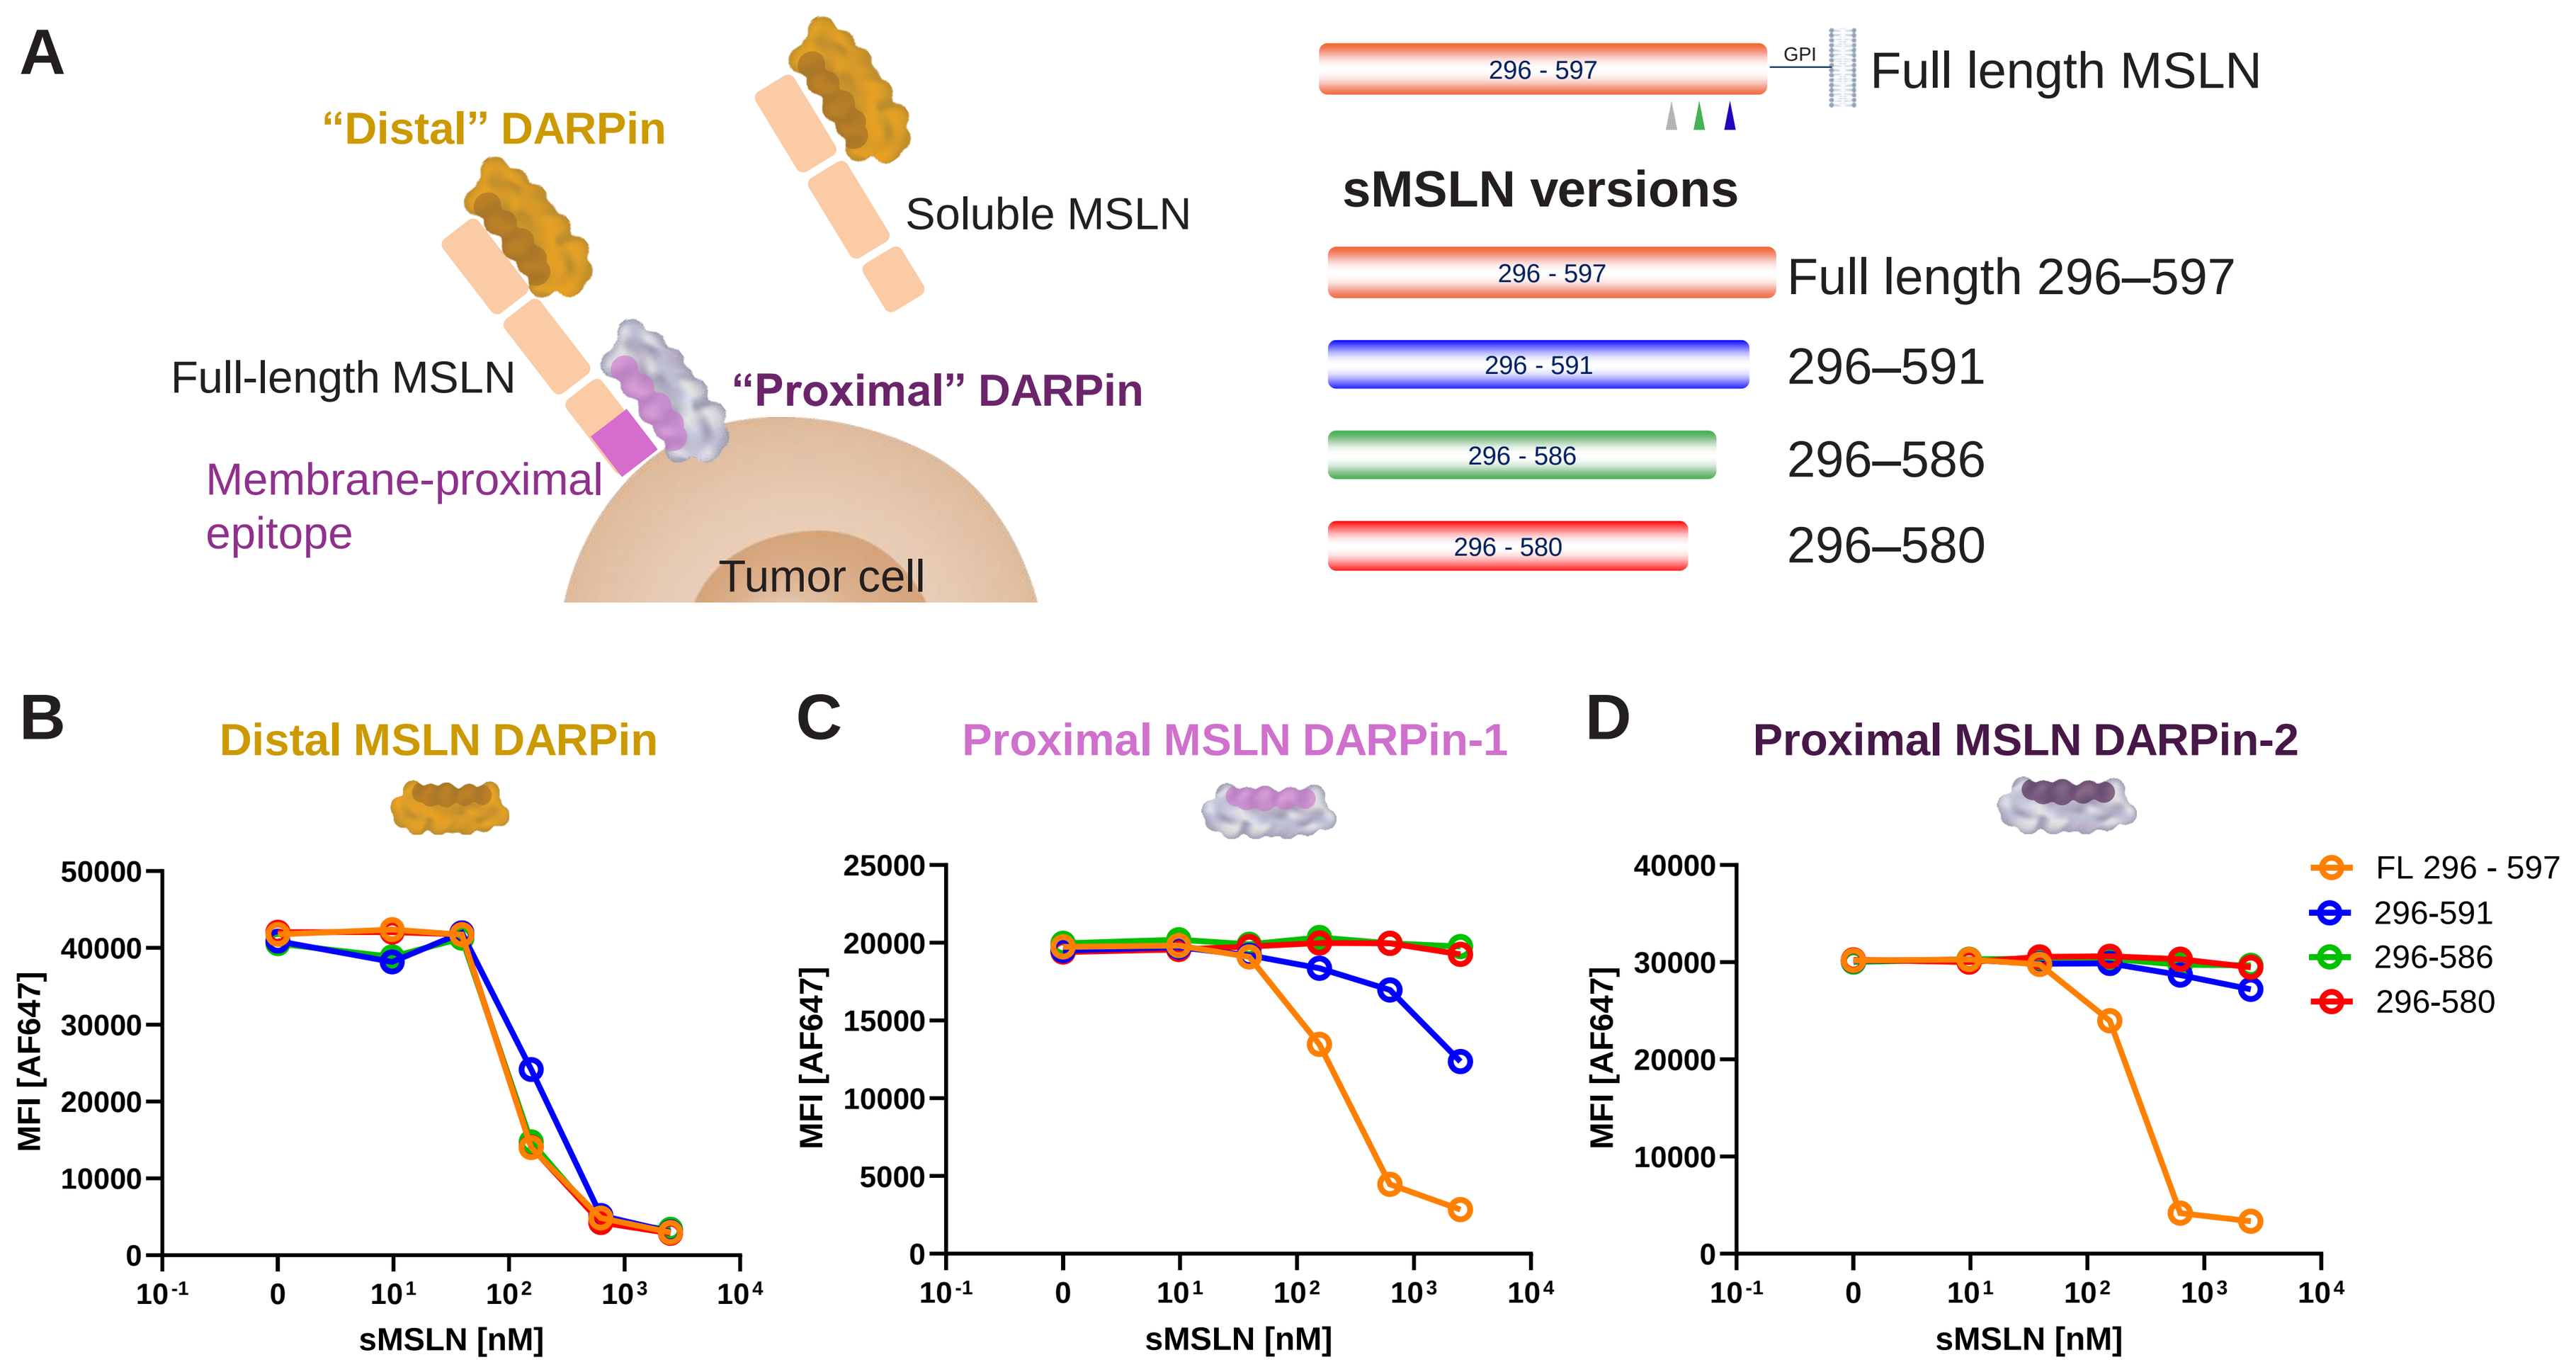
A
"Distal" DARPin
Soluble MSLN
Full-length MSLN
"Proximal" DARPin
Membrane-proximal epitope
Tumor cell
296 - 597
GPI
Full length MSLN
sMSLN versions
Full length 296–597
296–591
296–586
296–580
B
Distal MSLN DARPin
C
Proximal MSLN DARPin-1
D
Proximal MSLN DARPin-2
MFI [AF647]
sMSLN [nM]
FL 296 - 597
296-591
296-586
296-580

Figure 3: Cell binding of distal and proximal MSLN DARPins
DARPin binding to MSLN-expressing tumor cells was assessed in the presence of sMSLN versions at different concentrations. A) Schematic depicting the binding of distal and proximal MSLN DARPins and the sMSLN versions tested in the cell binding assay. Physiological major cleavage sites (after amino acid 584, 586 and 591) are indicated by the triangles.[4] B) Cell binding of a distal MSLN DARPin and two proximal MSLN DARPins C) and D) in presence of the indicated sMSLN versions. DARPins were detected with an anti-DARPin antibody, and the mean fluorescent intensity (MFI) values are indicated.

Favorable biodistribution of proximal MSLN DARPins

Biodistribution and tumor accumulation in hMSLN-MC38 model

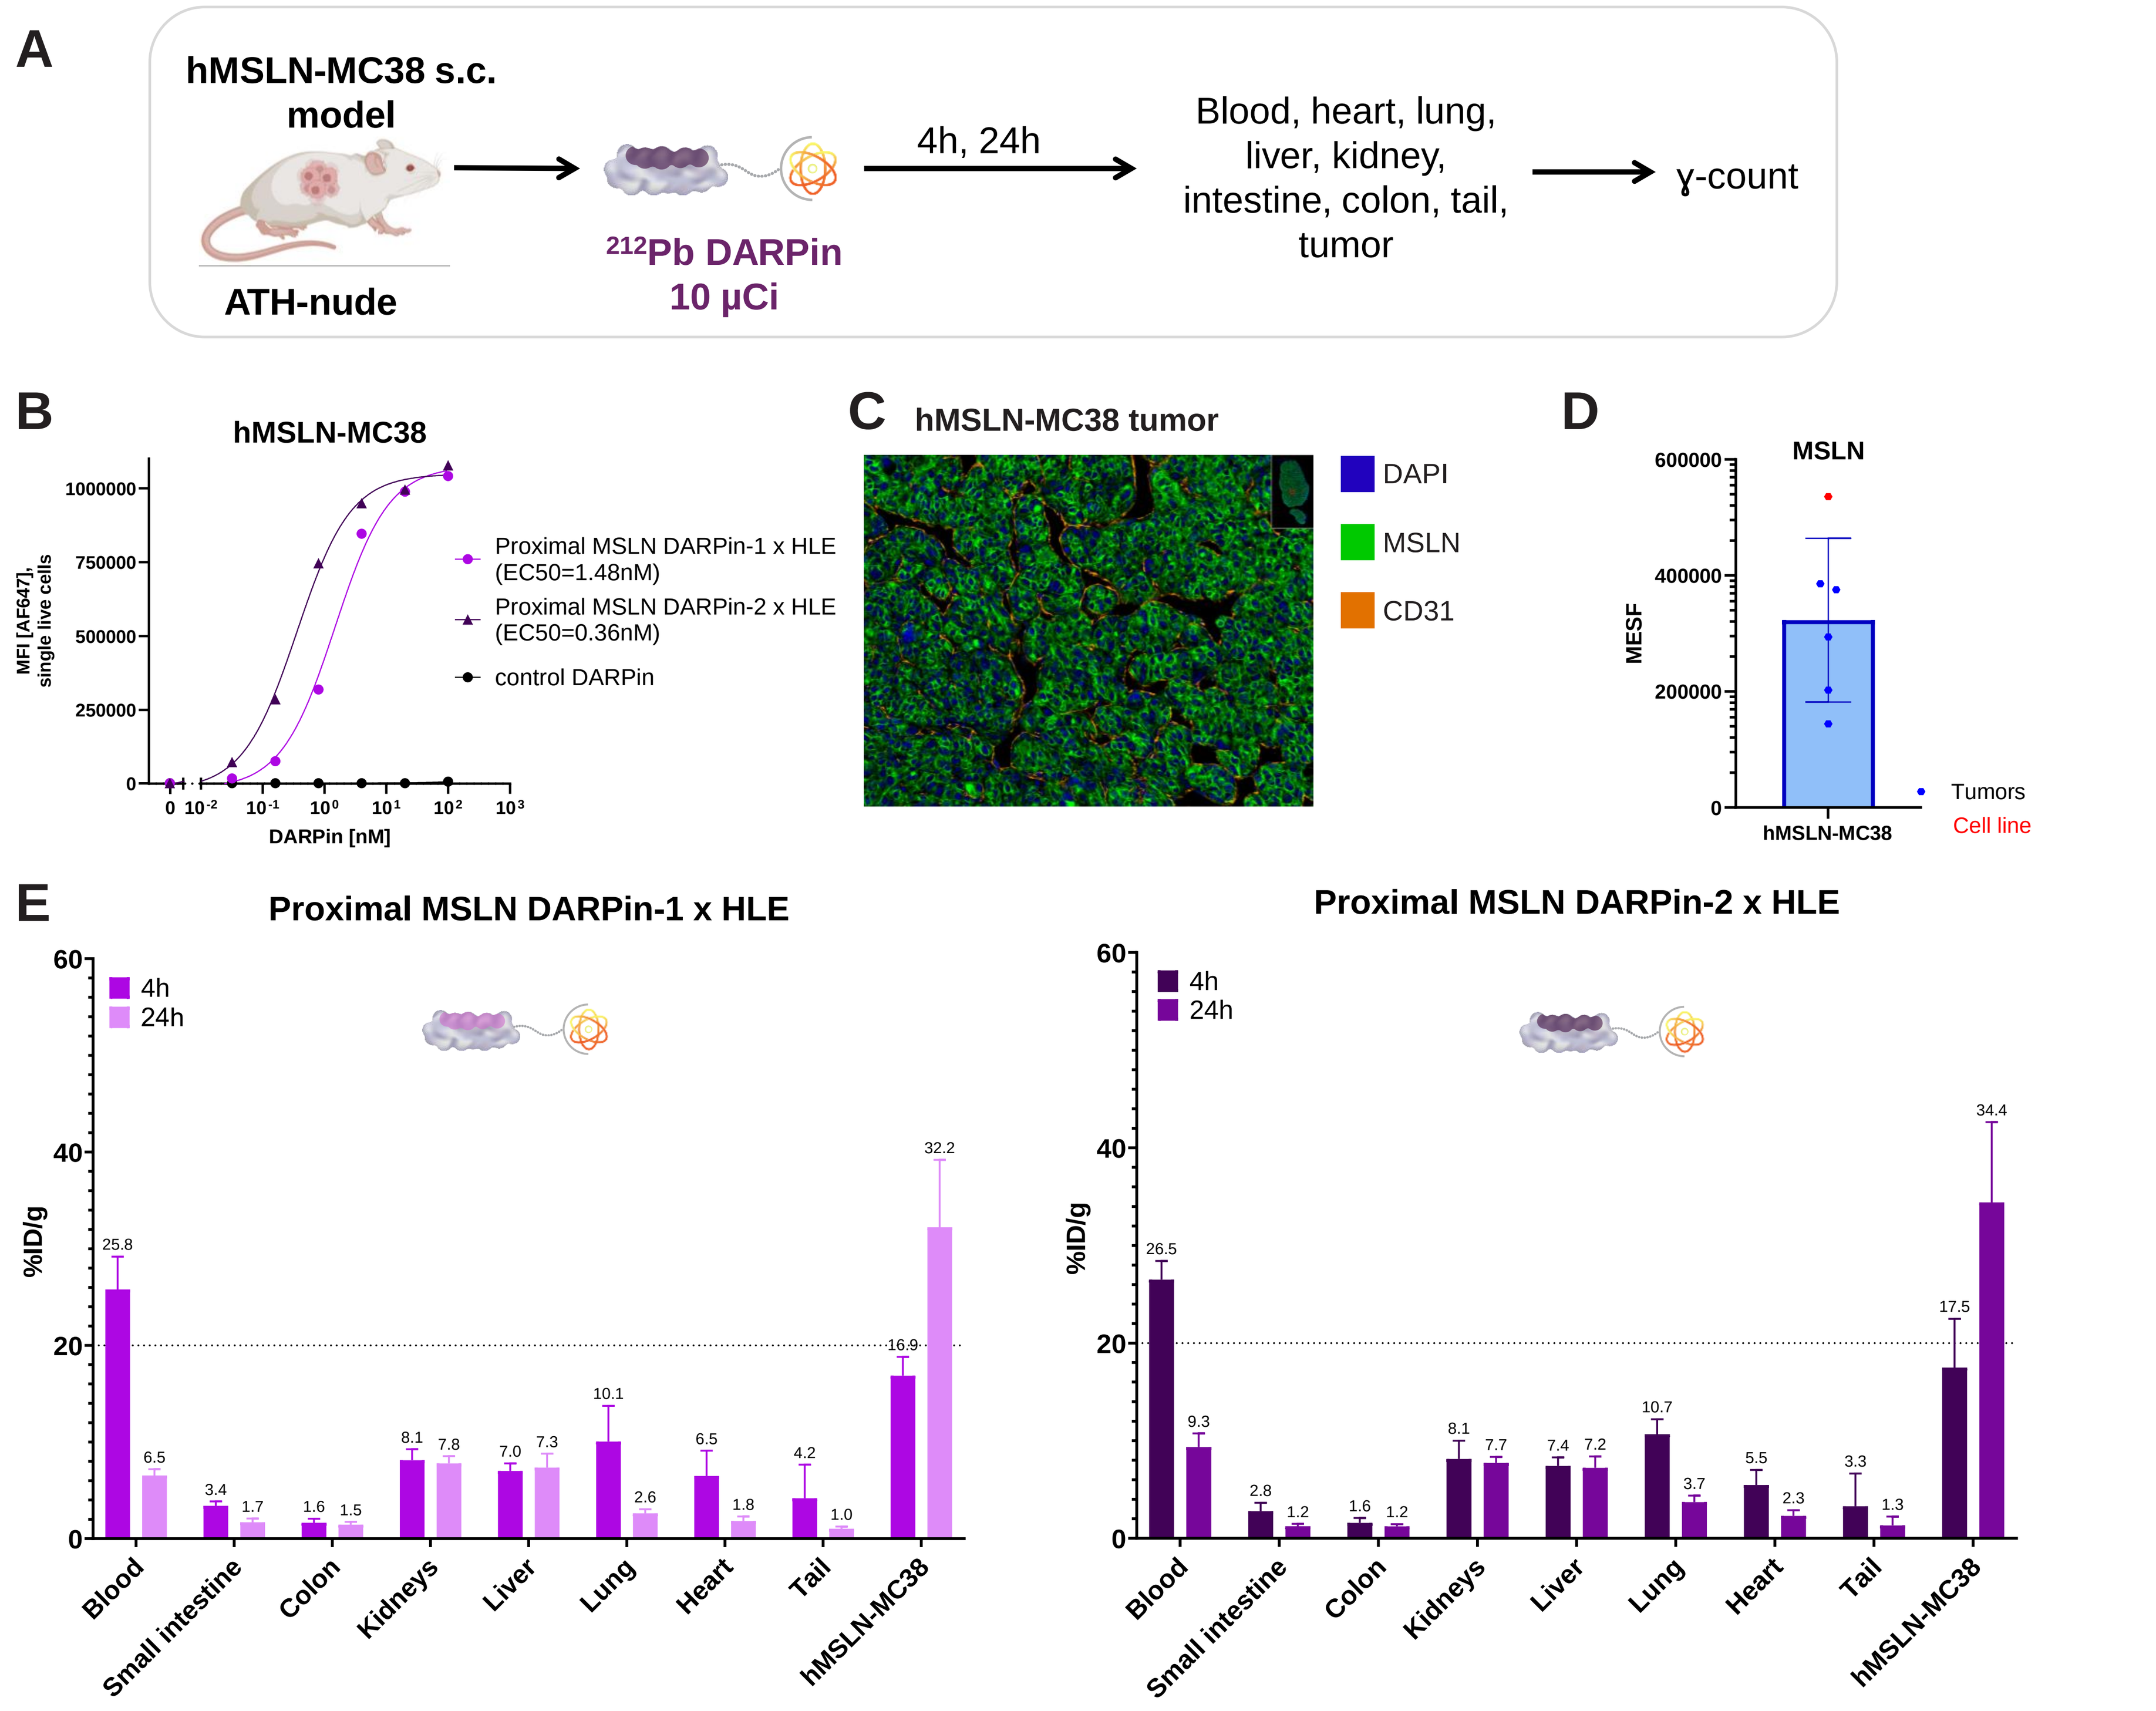
A
hMSLN-MC38 s.c. model
ATH-nude
212Pb DARPin
10 µCi
4h, 24h
Blood, heart, lung, liver, kidney, intestine, colon, tail, tumor
γ-count
B
hMSLN-MC38
Proximal MSLN DARPin-1 x HLE (EC50=1.48nM)
Proximal MSLN DARPin-2 x HLE (EC50=0.36nM)
control DARPin
DARPin [nM]
C
hMSLN-MC38 tumor
DAPI
MSLN
CD31
D
MSLN
MESF
hMSLN-MC38
Tumors
Cell line
E
Proximal MSLN DARPin-1 x HLE
Proximal MSLN DARPin-2 x HLE
4h
24h
%ID/g
Blood
Small intestine
Colon
Kidneys
Liver
Lung
Heart
Tail
hMSLN-MC38

Figure 4: Biodistribution of proximal MSLN DARPins in hMSLN-MC38 tumor model
A) Study design in hMSLN-MC38 model. Tumor bearing mice were administered with ^{212}Pb-labelled DARPins (10 µCi, 0.01 mg/kg) and tissue was collected for analysis 4 and 24 h post injection. B) Cell binding assay of proximal MSLN DARPins and control DARPin on hMSLN-MC38 cells. EC50 values are shown. C) MSLN and CD31 immunofluorescence staining of a hMSLN-MC38 tumor. Nuclei were visualized by DAPI. D) Flow cytometry analysis of MSLN expression in hMSLN-MC38 tumors (blue) and cell line (red). MSLN expression is shown as Molecules of Equivalent Soluble Fluorochrome (MESF). E) Biodistribution of Proximal MSLN DARPin-1 x HLE (left) and Proximal MSLN DARPin-2 x HLE (right) in the corresponding tissue shown as %ID/g at 4 and 24 h (n=5 mice/time point).

Conclusion

- MSLN is a promising target for ovarian cancer but high levels of sMSLN could sequester biotherapeutics and thus reduce their efficacy
- We developed a ^{212}Pb-based RDT with high affinity to membrane-proximal MSLN
- Binding of proximal MSLN DARPins to tumor cells *in vitro* was not affected by sMSLN
- *In vivo* results show a favorable biodistribution with strong tumor accumulation of ^{212}Pb x MSLN RDT in hMSLN-MC38 tumor bearing mice and modest accumulation in other organs
- These initial promising preclinical data provide the basis for further development efforts currently ongoing

Acknowledgments: Marcel Walser, Alienor Auge, Norbert Fic, Waleed Ali, Martin Hänggi, Quy Nguyen, Bradley Himes, Varaidzo Makina, Emily Brocious, Alyssa De Sisto, Caroline Crisp, Daniela Tapia Hernandez, Abigail Fondren, Angelina Roehrig
Conflict of interest disclosures: SW, DB, SR, MP, MM, CH, JW, DV, YK, NP, HF, TL, ET, JB, LA, CI, AC, CD, AG, and DS are employees and stock owners of Molecular Partners; ASa, TS, AW, ASc, and JT are employees of Orano Med.

For any questions, please contact: info@molecularpartners.com / attention of S. Wullschleger.

References: 1. Weidemann S, et al. Appl Immunohistochem Mol Morphol 2023;31:77–83, 2. Beyer HL, et al. Clin Chem 2007;53,666–72; 3. Hassan R, et al. Clin Cancer Res 2006;12:447–53; 4. Liu X, et al. PNAS 2022;119(19):e2202439119.